#369

Clinical and Pharmacokinetic/Pharmacodynamic Insights from CORE-008 Cohort A: Phase 2 Study of Intravesical Cretostimogene Grenadenorepvec in Patients with High-Risk BCG-Naïve Non-Muscle Invasive Bladder Cancer

Siamak Daneshmand, MD;[1] Colin P. N. Dinney, MD;[2*] Shane M. Pearce, MD[3] Laurence H. Belkoff, DO;[4*] Brian C. Mazzarella, MD;[5*] Christopher M. Pieczonka, MD;[6*] Neal D. Shore, MD;[7*] R. Jonathan Henderson, MD;[8*] Gary D. Steinberg, MD;[9*] and Trinity J. Bivalacqua, MD, PhD[10*]

[1] Keck School of Medicine of USC, Los Angeles, CA; [2] University of Rochester Medical Center, Rochester, NY; [3] Spokane Urology, Spokane, WA; [4] MidLantic Urology, Bala Cynwyd, PA; [5] Texas Oncology, Austin, TX; [6] Associated Medical Professionals of NY, Syracuse, NY; [7] Carolina Urologic Research Center, Myrtle Beach, SC; [8] Arkansas Urology, Little Rock, AR; [9] GDS Consulting, Chicago, IL; [10] Perelman School of Medicine at the University of Pennsylvania, Philadelphia, PA

For individual reference only. The information accessed through this QR code is intended solely for individual reference and should not be altered, modified, or reproduced in any way.

BACKGROUND

- Guidelines recommend IVE BCG or RC for HR NMIBC[1,2]
- IVE BCG challenges: variable durability across literature, treatment-limiting side effects, ongoing supply shortages[3-9]
- RC carries substantial morbidity and complications[10,11]
- Cretostimogene is an oncolytic immunotherapy with dual mechanisms of action that selectively replicates in and lyses cancer cells while simultaneously amplifying the immune response against bladder tumors
- CORE-008 is a phase 2, multi-arm, multi-cohort trial to evaluate the safety and efficacy of cretostimogene across a broad HR NMIBC population

METHODS

- Cohort A includes HR BCG-Naïve NMIBC with CIS (no prior BCG, BCG >24 months ago)
- Randomized 1:1:
 - Original (5-step): saline wash → DDM wash→ DDM dwell → saline wash→ cretostimogene dwell
 - Optimized (2-step): DDM wash→ cretostimogene dwell
- Response assessments: urine cytology, serial cystoscopy with directed biopsy, axial imaging (as indicated)
- Primary endpoint: CR at any time

Study Administration Schedule

	Induction	Maintenance/Follow-up							Ext. Maintenance/ Long-term Follow-up	
Month	0	3	6	9	12	15	18	21	24	30/36
Instillation	1 2 3 4 5 6	1 2 3(4 5 6)	1 2 3	1 2 3	1 2 3		1 2 3		1 2 3	(1 2 3)

RESULTS

- Majority of patients are: Male (90.7%), White (92.6%), >65 years (88.8%)
- As of Sept 01, 2025: Median follow-up 4.6 months; 83.7% overall CR
- Consistent treatment effect across patient subgroups
- No patients required radical cystectomy
- No treatment-related progression to MIBC or mUC; 3 patients experienced NMIBC stage reclassification
- 0% Grade ≥ 3 TRAEs, SAEs or deaths; No tx-related discontinuations

CR Rate	% (n/N)	95% CI
Original Admin **(5-Step)**	79.2% (19/24)	(57.8, 92.9)
Optimized Admin **(2-Step)**	88.0% (22/25)	(68.8, 97.5)

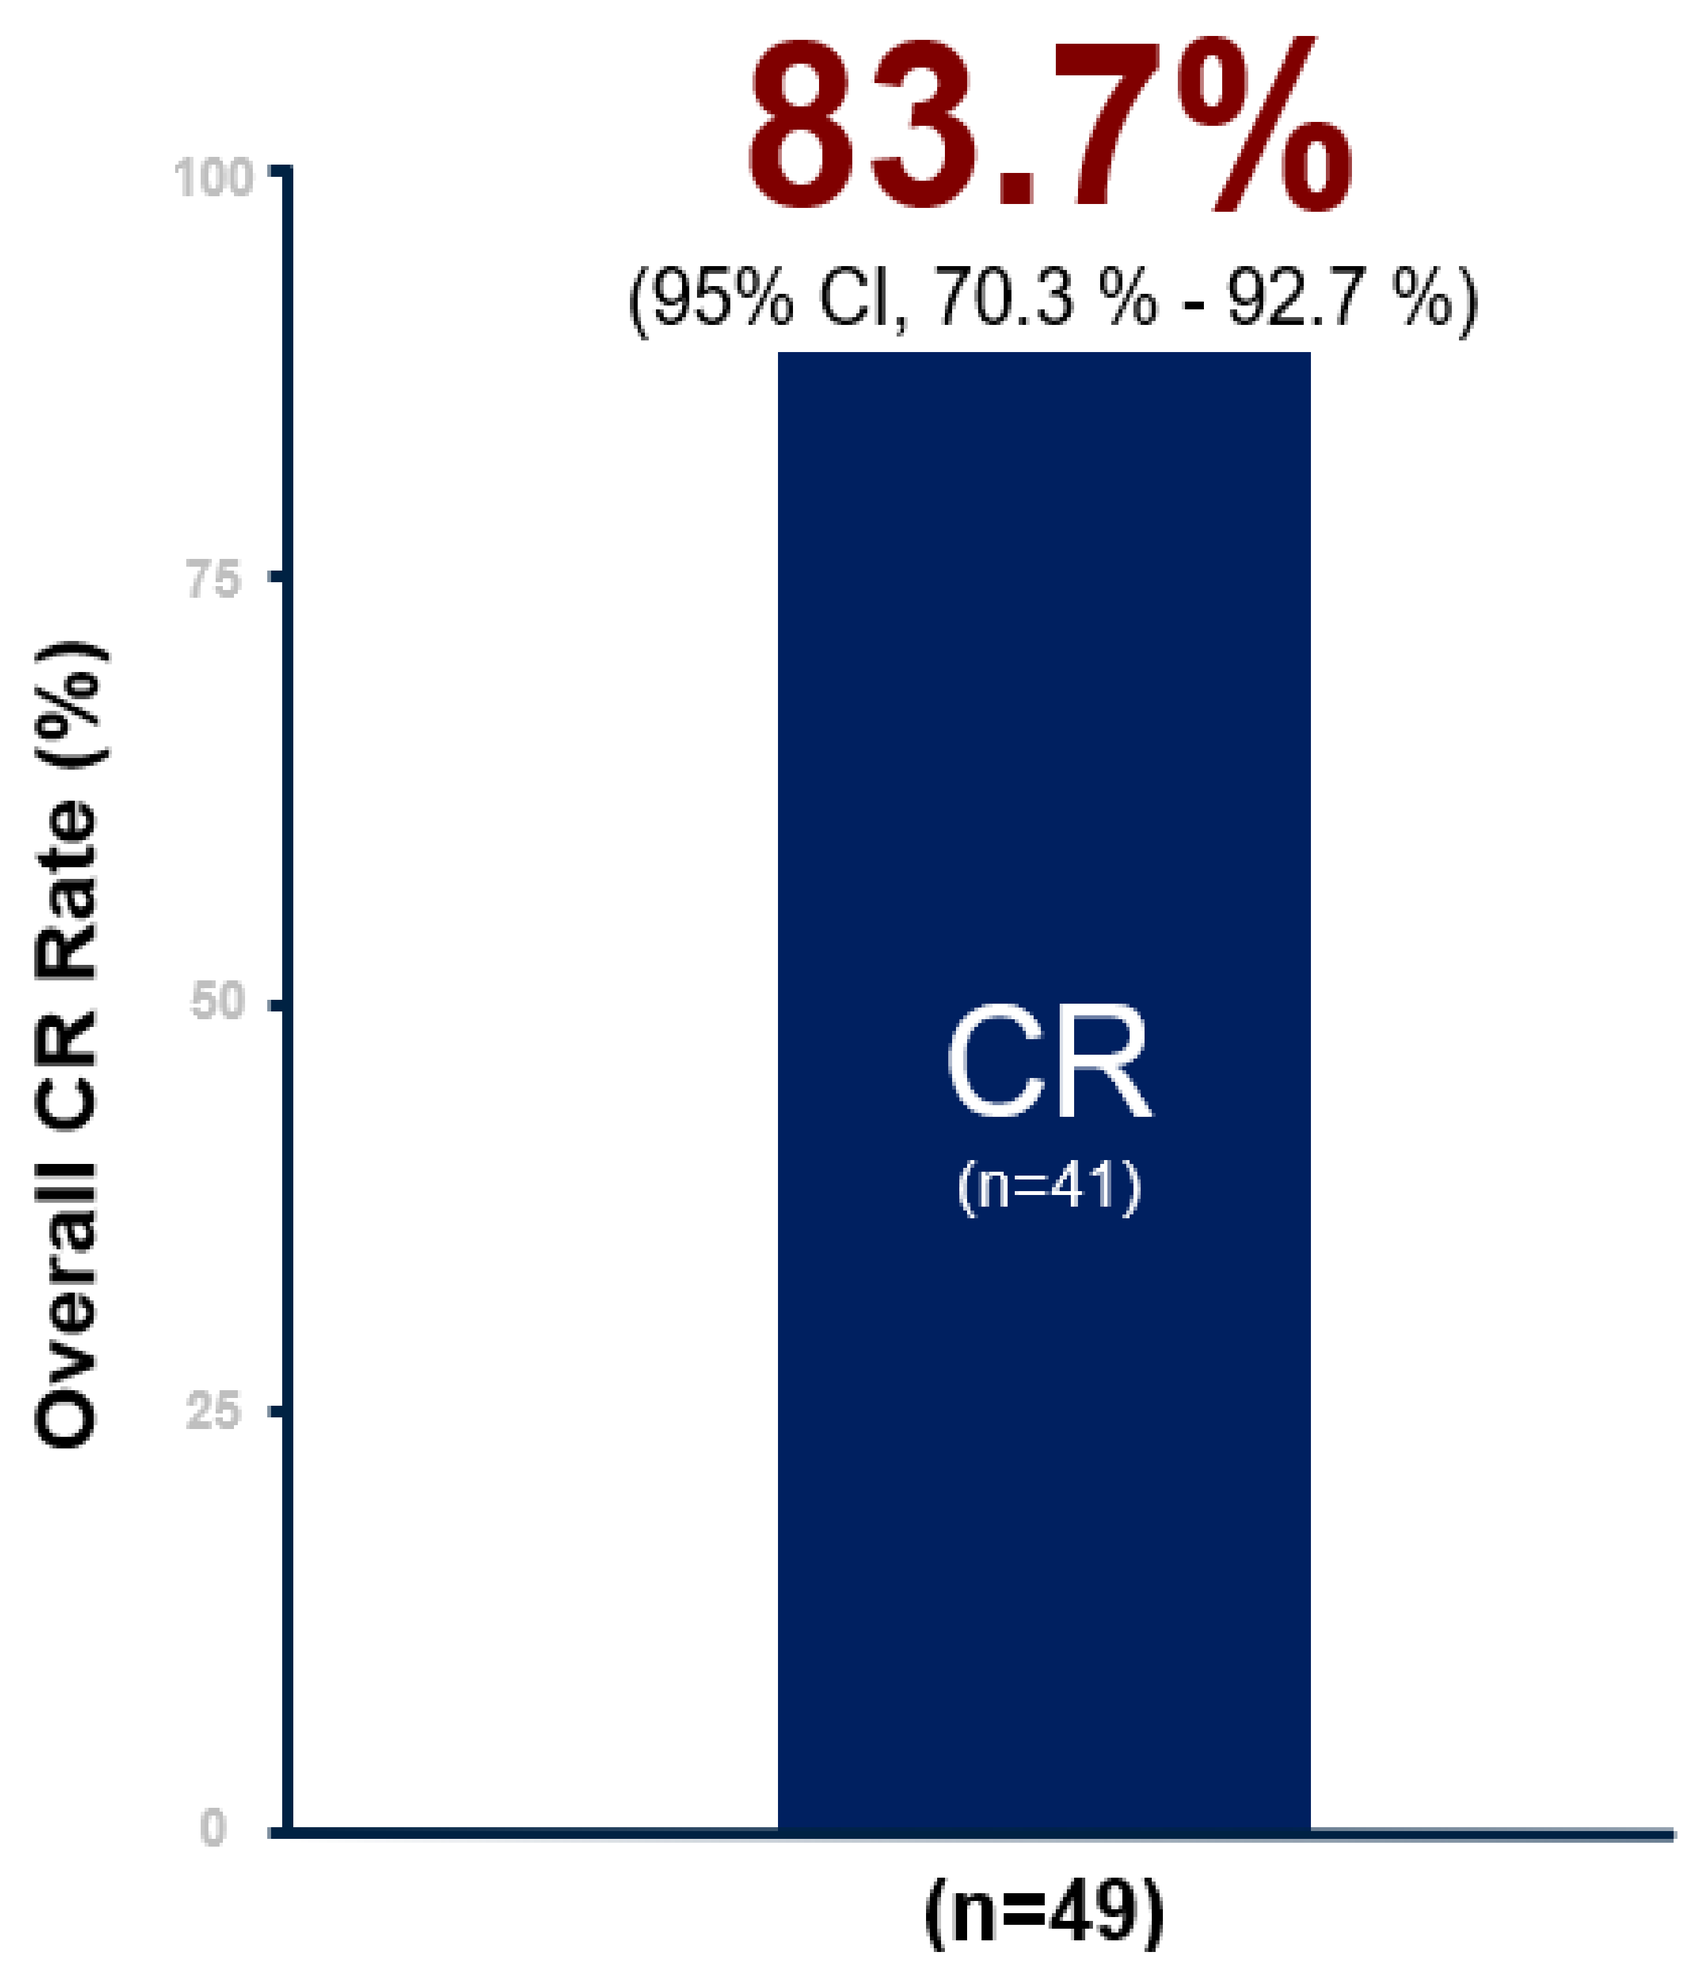
83.7%
(95% CI, 70.3 % - 92.7 %)
Overall CR Rate (%)
100
75
50
25
0
CR
(n=41)
(n=49)

Initial, Clinical Results in BCG-Naïve NMIBC

Translational Insights Reinforce Cretostimogene's MOA, Treatment Schedule, and Safety Profile

RESULTS

Cretostimogene Replication in Urine

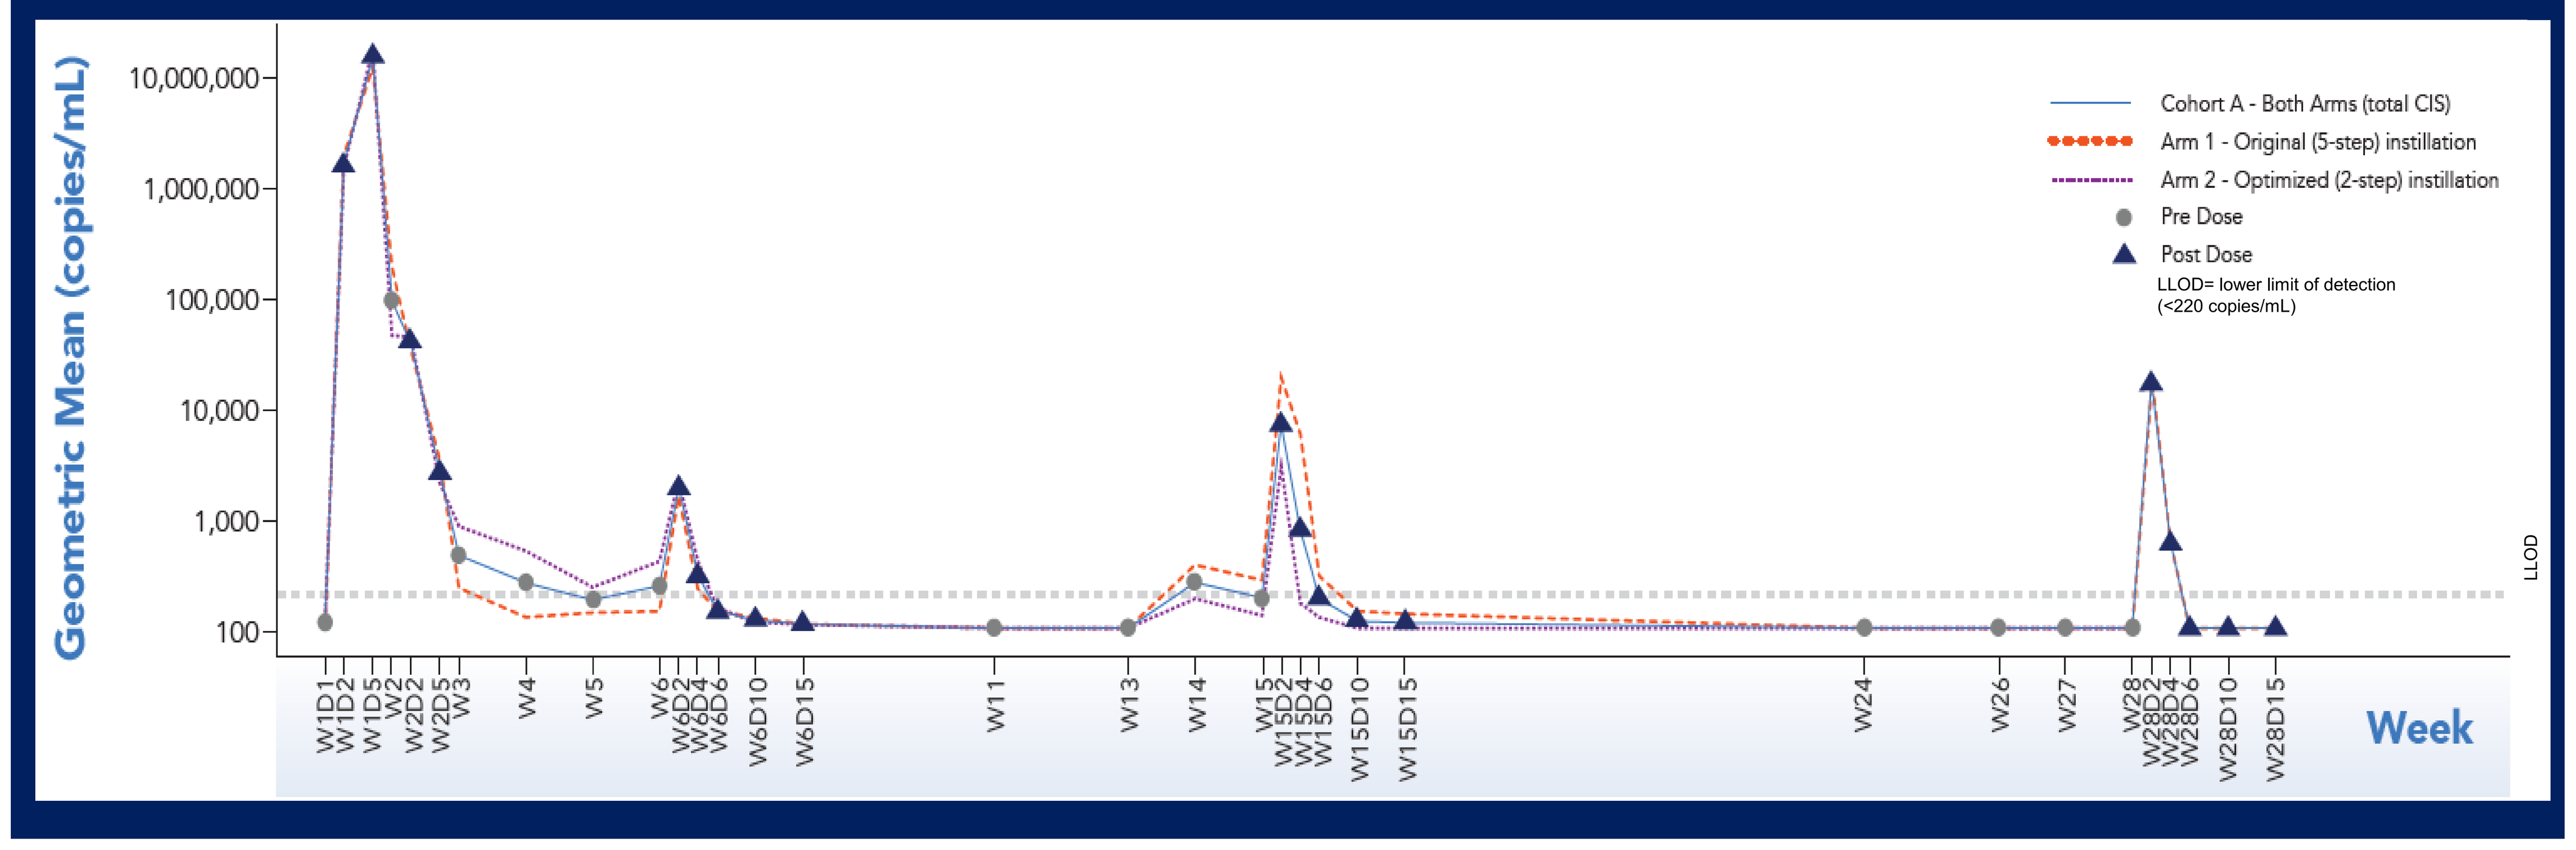
Geometric Mean (copies/mL)
10,000,000
1,000,000
100,000
10,000
1,000
100
Cohort A - Both Arms (total CIS)
Arm 1 - Original (5-step) instillation
Arm 2 - Optimized (2-step) instillation
Pre Dose
Post Dose
LLOD= lower limit of detection (<220 copies/mL)
LLOD
W1D1 W1D2 W1D5 W2 W2D2 W2D5 W3 W4 W5 W6 W6D2 W6D4 W6D6 W6D10 W6D15 W11 W13 W14 W15 W15D2 W15D4 W15D6 W15D10 W15D15 W24 W26 W27 W28 W28D2 W28D4 W28D6 W28D10 W28D15
Week

- No detectable cretostimogene levels and low endogenous GM-CSF in the urine of the 53 patients' samples tested at baseline
- Transient increase in cretostimogene levels during induction, declining below the limit of detection after induction and maintenance dosing, with no evidence of latent or extended replication
- No appreciable differences in viral levels between arms
- Consistent with findings from BOND-003 Cohort C (BCG-UR with CIS)
- Effective viral clearance supports relaxed close-contact precautions

GM-CSF Transgene Expression in Urine

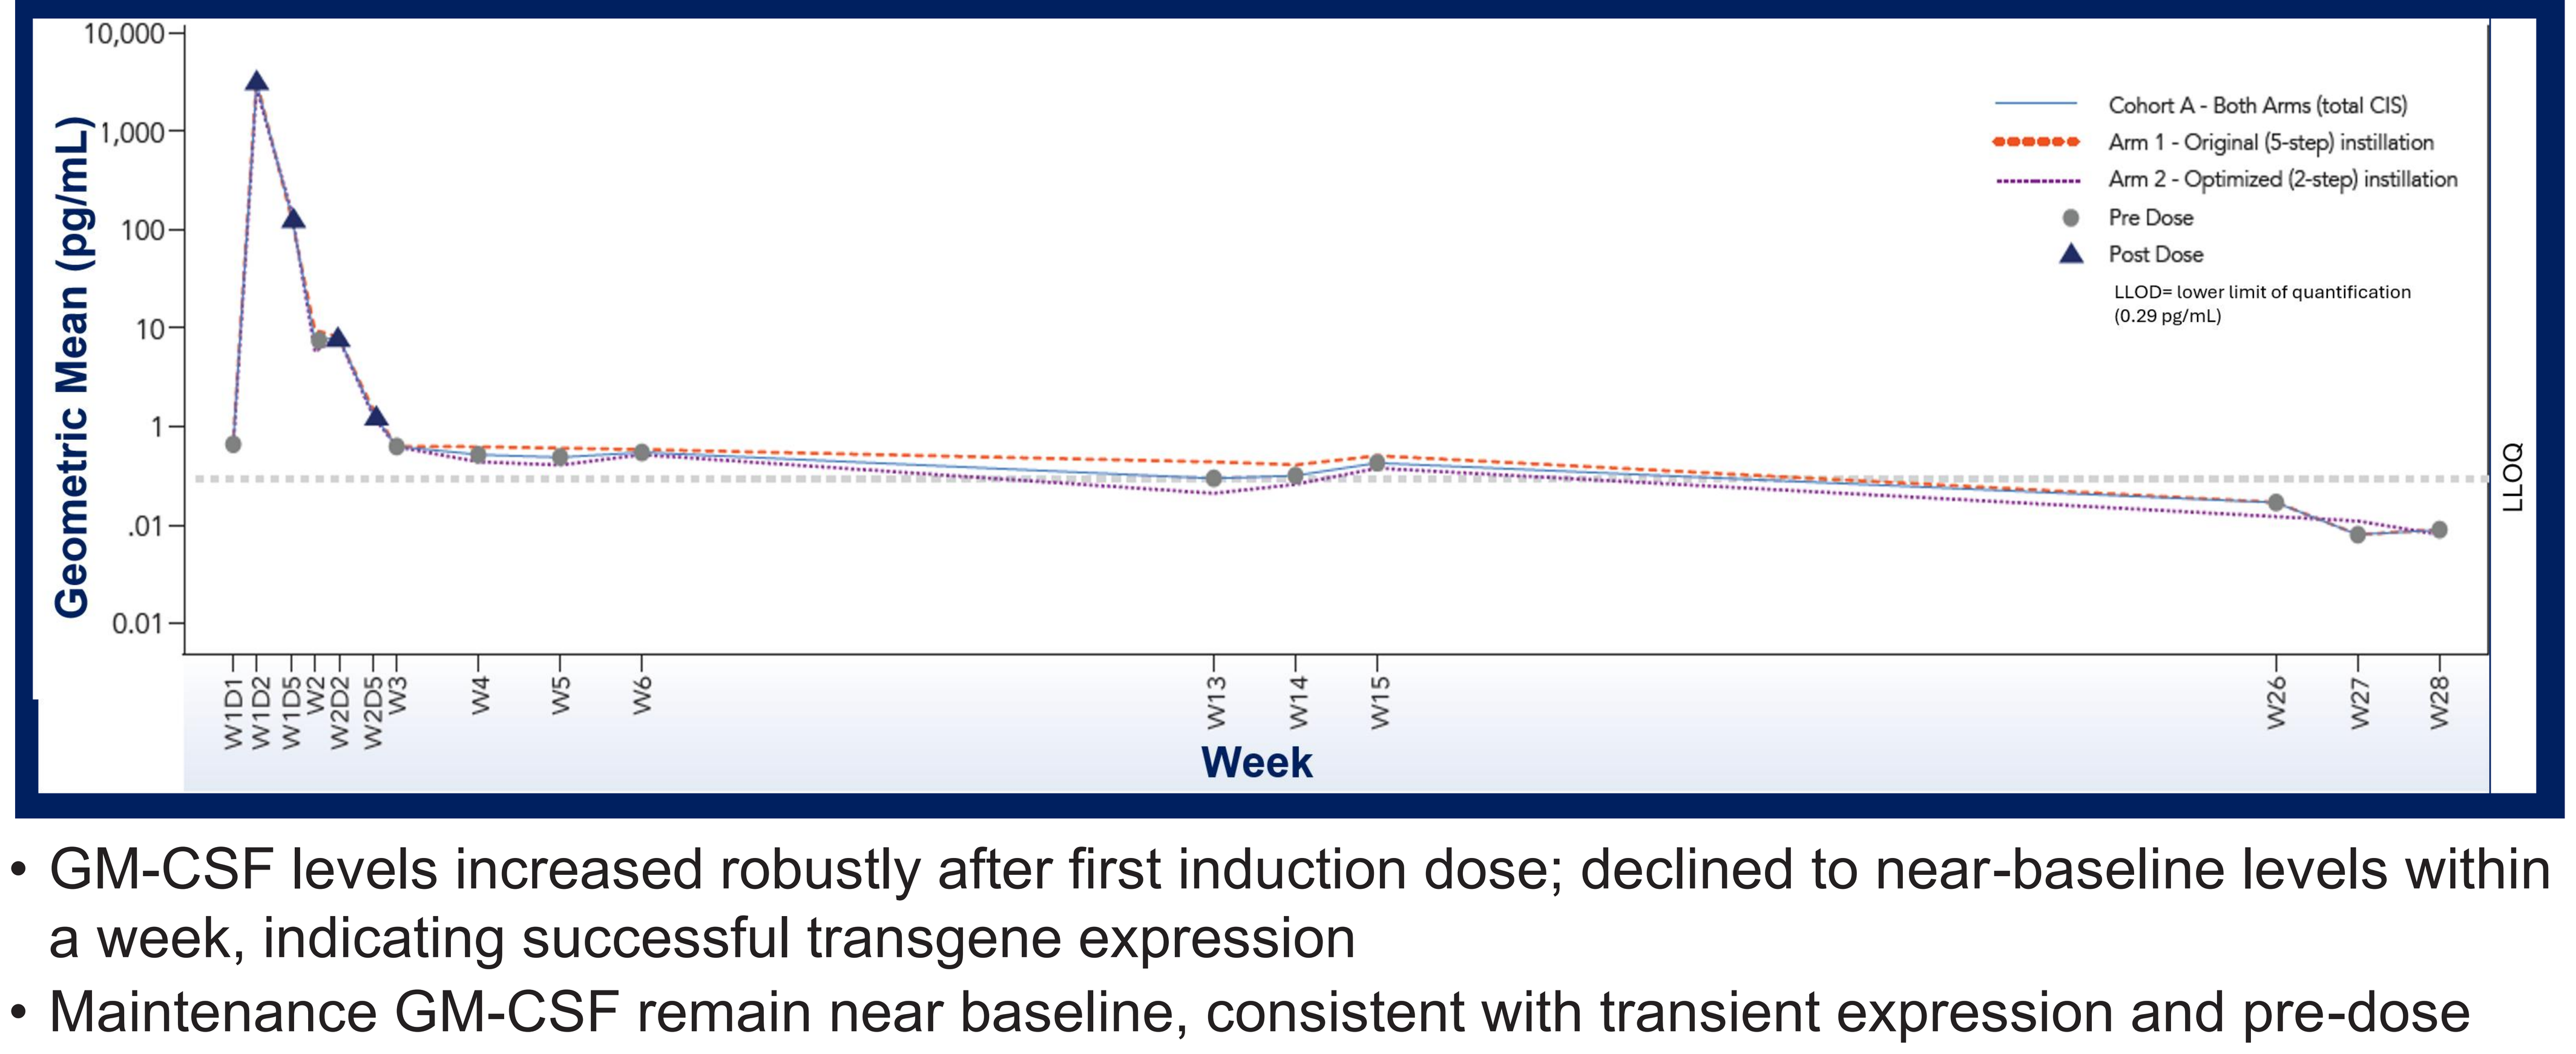
Geometric Mean (pg/mL)
10,000
1,000
100
10
1
.01
0.01
Cohort A - Both Arms (total CIS)
Arm 1 - Original (5-step) instillation
Arm 2 - Optimized (2-step) instillation
Pre Dose
Post Dose
LLOD= lower limit of quantification (0.29 pg/mL)
LLOQ
W1D1 W1D2 W1D5 W2 W2D2 W2D5 W3 W4 W5 W6 W13 W14 W15 W26 W27 W28
Week

- GM-CSF levels increased robustly after first induction dose; declined to near-baseline levels within a week, indicating successful transgene expression
- Maintenance GM-CSF remain near baseline, consistent with transient expression and pre-dose sampling
- No substantial differences in GM-CSF levels between arms
- Transient GM-CSF expression aligns with intended MOA

Abbreviations: IVE: Intravesical; BCAN: Bladder Cancer Advocacy Network; CR: complete response; DoR: duration of response; HG: high-grade; HR: high-risk; NMIBC: non-muscle invasive bladder cancer; RC: radical cystectomy; MIBC: muscle invasive bladder cancer; mUC: metastatic urothelial cancer; SAE: serious adverse event; TRAE: treatment-related adverse event

References: **1** NCCN Bladder Cancer Guidelines; 2026, **2** AUA/SUO NMIBC Guidelines; 2026 **3** Sylvester,*Eur Urol*; 2006, **4** Kamat,*J Clin Oncol*; 2016, **5** Roumiguie,*Eur Urol*; 2022, **6** Longoni,*Eur Urol Oncol*; 2025, **7** Brausi,*Eur Urol,* 2014, **8** Tapiero,*Urology,*2018, **9** Van Der Meijen,*Eur Urol* 2003, **10** Maibom,*BMJ Open,* 2021, **11** Clements,*Eur Urol* 2021

Acknowledgements: Sarah Donatelli, PhD; Kirk Keegan, MD; Rob Svatek, MD; Rebecca Tregunna, MD, MBA; Shelja Patel, PharmD; Sarah D. Hohl, PhD; and Vijay Kasturi, MD

Contact Information: Siamak Daneshmand, MD: daneshma@med.usc.edu

#369

Clinical and Pharmacokinetic/Pharmacodynamic Insights from CORE-008 Cohort A: Phase 2 Study of Cretostimogene in Patients with High-Risk BCG-Naïve NMIBC

- Patients with HR BCG-Naïve NMIBC with CIS randomized:
 - Original (5-step) administration
 - Optimized (2-step) administration
- Overall CR: 83.7% (95% CI: 70.3, 92.7)
- 90.1% completed all protocol-defined treatments (Optimized admin: 100%)
- 0 grade ≥ 3 TRAEs
- GM-CSF levels increased robustly after first induction dose, indicating successful transgene expression
- No detectable cretostimogene and low endogenous GM-CSF

01Sept2025 data cut off

Cretostimogene Shedding in Urine

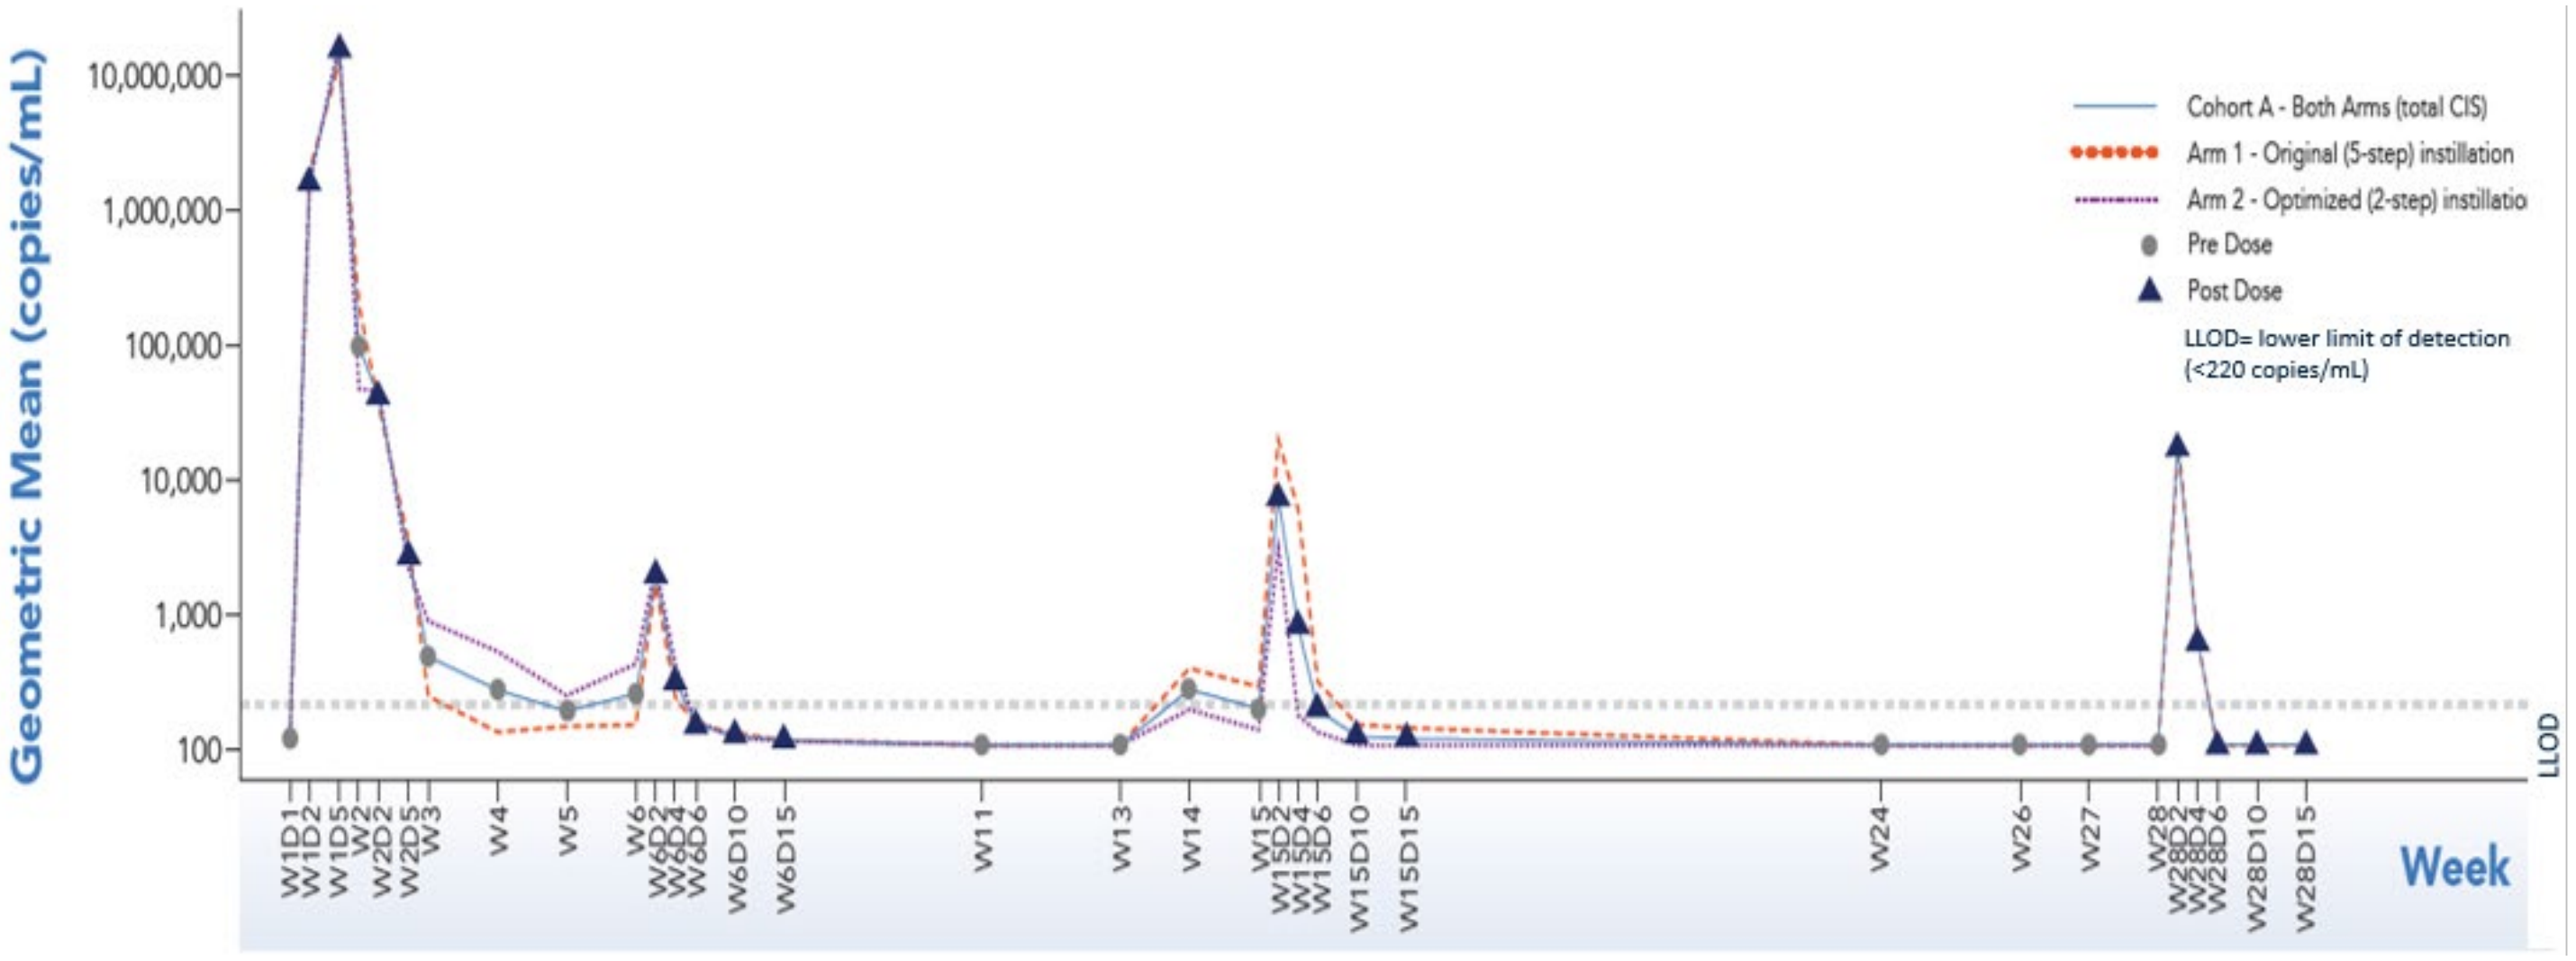

Geometric Mean (copies/mL)
10,000,000
1,000,000
100,000
10,000
1,000
100
Cohort A - Both Arms (total CIS)
Arm 1 - Original (5-step) instillation
Arm 2 - Optimized (2-step) instillatio
Pre Dose
Post Dose
LLOD= lower limit of detection
(<220 copies/mL)
LLOD
W1D1 W1D2 W1D5 W2 W2D2 W2D5 W3 W4 W5 W6 W6D2 W6D4 W6D6 W6D10 W6D15 W11 W13 W14 W15 W15D2 W15D4 W15D6 W15D10 W15D15 W24 W26 W27 W28 W28D2 W28D4 W28D6 W28D10 W28D15
Week

CR Rate	% (n/N)	(95% CI)
Original 5-step	**79.2%** *19/24 patients*	(57.8, 92.9)
Optimized 2-step	**88.0%** *22/25 patients*	(68.8, 97.5)

For individual reference only. The information accessed through this QR code is intended solely for individual reference and should not be altered, modified, or reproduced in any way.

USC Department of Urology